A UNIQUE SENSITIVE SKIN CLEANSING TECHNOLOGY

DG Kern[1], ZD Draelos[2], J Namkoong[1], M Riggs[1], HE Knaggs[1]

[1]Center for Anti-Aging Research, Nu Skin Enterprises, Inc., Provo, UT
[2]Dermatology Consulting Services, High Point, NC

BACKGROUND

Sensitive skin is challenging to diagnose since there are different chemical, environmental and psychological triggers and sometimes without visual symptoms. In the first epidemiological study, 51.4% of the women and 38.2% of the men claimed to have sensitive skin [1]. The etiology of sensitive skin is unknown, but some studies suggest the epidermal barrier impairment as a cause of sensitive skin [2]. Shear force and/or friction during cleansing can induce skin damage [3]. Cleansing motion with potential chemical triggers can magnify the symptoms of sensitive skin by inducing barrier damage, yet the skin of afflicted individuals must be maintained in a hygienic condition. In order to improve the skin cleanliness without damage, a novel technology was developed to target sensitive skin individuals.

OBJECTIVE

1. To further the development of a mechanically-moved polymer surface of unique topography to provide treatment and gentle cleansing
2. To assess possible differences in treatment/cleansing surface topographies in cleansing sensitive skin.

METHODS

This IRB-approved clinical research study enrolled 16 female subjects, 25-65 years of age, Fitzpatrick skin types I-II, with self-assessed sensitive skin confirmed by the dermatologist investigator in a panel comprised of subjects with rosacea, eczema, atopic dermatitis or cosmetic intolerance syndrome. Subjects used the unique cleansing device constructed to provide a counter-oscillating movement (Figure 1) of a cleansing surface composed of a soft polymer of unique topography and a commercially-available sensitive skin cleanser twice daily for 14 days (Photo 1) Subjects' facial skin was evaluated by self-assessment and clinician grading at baseline and days 7 and 14 both before and after cleansing. A Mann-Whitney two-tailed paired t test was used to analyze the non-parametric data. The device data was compared to baseline longitudinally for the primary treatment/cleansing surface. A separate analysis was conducted for the secondary treatment/cleansing surface for those who experienced irritation with the primary surface.

CONFLICT OF INTEREST

DGK, JN, MR and HEK are employees of Nu Skin Enterprises, Inc. ZDD received funding from Nu Skin Enterprises, Inc.

RESULTS

The investigator noted immediate improvement following one cleansing in skin smoothness (p=0.009), softness (p=0.017), texture (p=0.028), and cleansing ability (p<0.001). Further sustained improvement occurred in all of these attributes, including pores, with all parameters being highly statistically significant (p<0.001) after 7 days of use with continued excellent performance until the study conclusion at day 14. The sensitive skin subjects noted improvement in skin softness (p=0.011) and smoothness (p=0.008) immediately after one use of the treatment/cleansing device. After 7 days of use, the subjects rated improvement in smoothness (p=0.002), softness (p=0.002), pores (p=0.041), texture (p=0.002), and cleansing ability (p=0.019). This improvement continued into day 14 post cleansing where highly statistically significant (p<0.001) improvement was seen in smoothness, softness, pores, and texture with statistically significant improvement in cleansing ability (p=0.002). All 16 subjects completed the study. No tolerability issues were noted by the investigator dermatologist or the sensitive skin subjects. Additional research is being conducted to evaluate this unique technology.

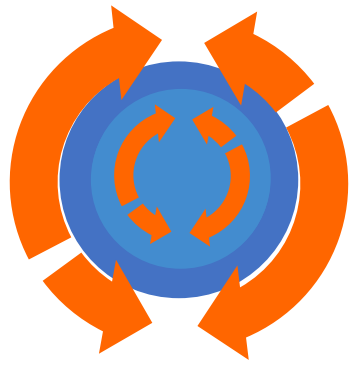

Figure 1. Representation of the counter oscillating motion of the soft-polymer surface.

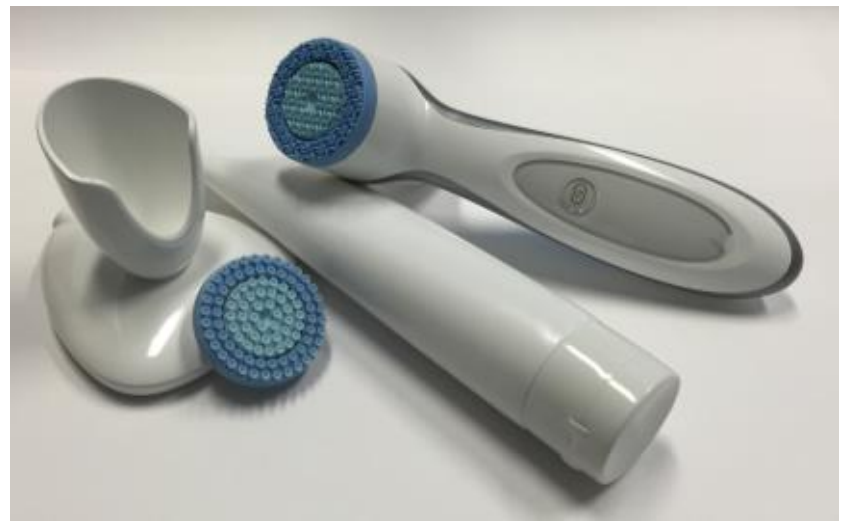

Photo 1. Soft-polymer, counter oscillating facial cleansing device.

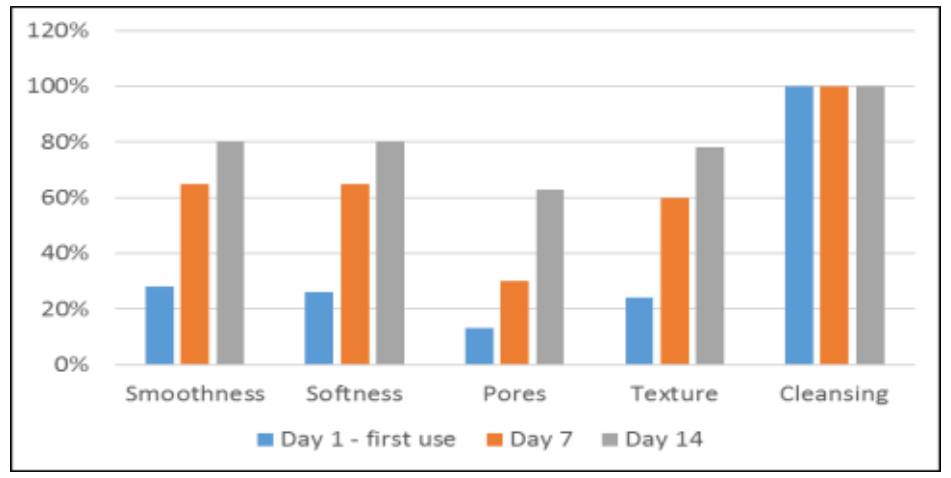
120%
100%
80%
60%
40%
20%
0%
Smoothness
Softness
Pores
Texture
Cleansing
Day 1 - first use
Day 7
Day 14

Figure 2. Clinical investigator assessment. Percent change over initial visit, untreated, using a 5-point facial attribute grading scale: 0=none, 1=minimal, 2= mild, 3= moderate, 4=severe. Assessments were performed at the research center 10 – 20 minutes after device use in clinic. After Day 7, all improvements were statistically significant (p<0.001).

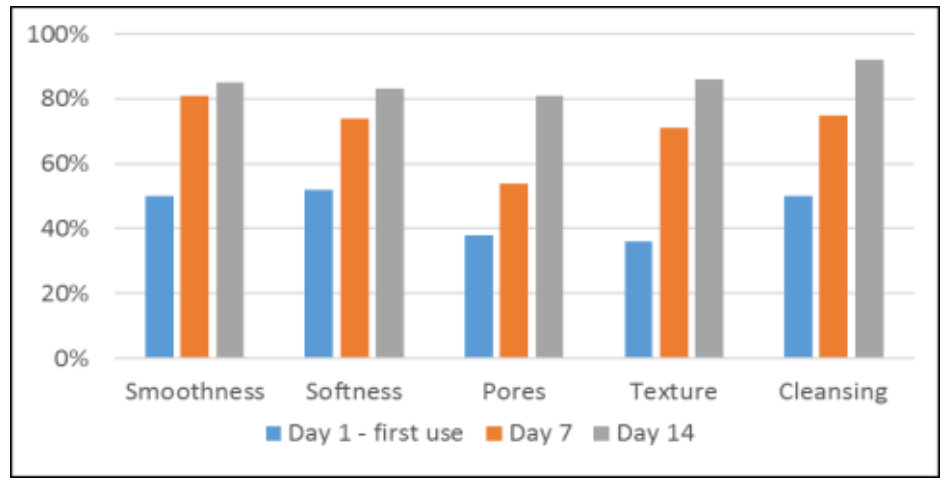
100%
80%
60%
40%
20%
0%
Smoothness
Softness
Pores
Texture
Cleansing
Day 1 - first use
Day 7
Day 14

Figure 3. Subject self-assessment. Percent change over initial visit, untreated, using a 5-point facial attribute grading scale: 0=none, 1=minimal, 2= mild, 3= moderate, 4=severe. Assessments were performed at the research center 10 – 20 minutes after device use in clinic. At Day 14, all improvements were statistically significant (p<0.001).

CONCLUSIONS

- A counter-oscillating unique soft polymer cleansing device provided aesthetic improvement and superior cleansing in subjects with sensitive skin.
- Agreement was seen between clinical investigator assessments and subject self-assessments confirming the technological benefit of a novel device-based treatment/cleansing regimen.

REFERENCES

1. Willis CM, Shaw S, DeLacharriere O, Baverel M, Reiche L, Jourdain R, Bastien P, Wilkinson JD. Sensitive skin: an epidemiological study. Br J Dermatol. 2001, 145: 258–263.
2. Misery, L., Loser, K. and Stander, S. Sensitive skin. J Eur Acad Dermatol Venereol. 2016, 30(S1): 2-8.
3. Koenig, DW, Dvoracek, B. and Vongsa, R. In vitro prediction of in vivo skin damage associated with the wiping of dry tissue against skin. Skin Research and Technology 2013, 19: e453–e458.